2768-LB

Evaluation of a nutrition program using a diabetes-specific formula with β-Hydroxy-β-Methylbutyrate (HMB) on metabolic and muscle health in patients with type 2 diabetes with or at risk of malnutrition.

J. M. Garcia-Almeida[1], J. M. Guardia-Baena[2], S. Palma Milla[3,4,5], M. Camprubi Robles[*6], G. Guzmán Rolo[7], G. E. Baggs[8], P. Suryawanshi[9], M. J. Martínez-Ramírez[10], M. J. Molina-Puerta[11], J. F. Merino-Torres[12,13], M. Civera-Andrés[14,15], P. P. Garcia-Luna[16], G. Olveira-Fuster[17,18,19], J. I. Botella-Carretero[20], I. Rebollo-Pérez[21], F. J. Vílchez-López[22,23], M. Maíz-Jiménez[24], Y. García-Delgado[25], L. A. Calles-Romero[26], P. Matía-Martín[27,28]

1. Endocrinology and Nutrition Service, Hospital Universitario Virgen de la Victoria, Málaga, 2. Endocrinology and Nutrition Service, Hospital Universitario Virgen de las Nieves, Granada, 3. Coordinator of the Clinical Nutrition and Dietetics Unit. Endocrinology and Nutrition Service, Hospital Universitario La Paz, 4. Associate Professor of Health Sciences, Department of Medicine, Universidad Autónoma de Madrid, 5. Instituto de Investigación Sanitaria IdiPAZ, Madrid, 6. Abbott Nutrition R&D, Abbott Laboratories, Granada, Spain 7. Medical Department, Abbott Laboratories, 8. Abbott Nutrition R&D, Abbott Laboratories, Columbus, OH, USA. 9. Statistical Services, Cognizant Technologies Solution Pvt. Ltd., Mumbai, India, 10. Head of Service and Director of UGC Endocrinology and Nutrition, Hospital Universitario de Jaén, Jaen, 11. Endocrinology and Nutrition Service, Hospital Universitario Reina Sofia, Córdoba, 12. Endocrinology and Nutrition Service, Hospital Universitario Politécnico La Fe, 13. Professor of Medicine, Universidad de Valencia, 14. Endocrinology and Nutrition Service, Hospital Clínico Universitario de Valencia, 15. INCLIVA Instituto de investigación Sanitaria, Valencia, 16. Endocrinology and Nutrition Clinical Management Unit, Instituto de Biomedicina de Sevilla (IBiS), Hospital Universitario Virgen del Rocío/CSIC/Universidad de Sevilla, Sevilla, 17. Universidad de Malaga, Instituto de Investigación Biomedica de Málaga (IBIMA)-Bionand Platform, 18. Endocrinology and Nutrition Department, Hospital Regional Universitario de Málaga, Málaga, 19. CIBER of Diabetes and Associated Metabolic Diseases, Instituto de salud Carlos III, 20. Endocrinology and Nutrition Department, Hospital Universitario Ramón y Cajal, Madrid, 21. Endocrinology and Nutrition Unit, Hospital Universitario Juan Ramón Jimenez de Huelva, Huelva, 22. Endocrinology and Nutrition Department, Hospital Universitario Puerta del Mar, 23. Instituto de Investigación e Innovación Biomédica de Cádiz, Cádiz, 24. Endocrinology and Nutrition Department, Hospital Universitario 12 de Octubre, Madrid, 25. Endocrinology and Nutrition Department, Complejo Hospitalario Universitario Insular-Materno Infantil, Gran Canaria, 26. Endocrinology and Nutrition Department, Hospital Universitario Basurto, Bilbao, Spain, 27. Endocrinology, Metabolism and Nutrition Department, Hospital Clínico San Carlos, 28. Instituto de Investigación Sanitaria del Hospital Clínico San Carlos (IdISSC), Madrid, Spain.

INTRODUCTION & OBJECTIVE:

In adults with **type 2 diabetes** (T2DM) Malnutrition (MN) and Sarcopenia are highly prevalent and are associated with adverse clinical outcomes.

STUDY GOAL

- Evaluate multi-modal nutritional support programme in outpatients with T2DM with/at risk of MN on standard of care
- Using a high-protein diabetes-specific nutritional formula with HMB, Arginine (Arg) and Lysine (Lys) (Glucerna® Advance (Abbott))
- On nutritional, glycaemic, functional and body composition outcomes

METHOD:

A PROSPECTIVE, REAL-WORLD EVIDENCE STUDY

in adults with T2DM and 'MUST' score ≥2 from 16 hospitals in Spain

INTERVENTION FOR 90 DAYS:

- Dietary and physical exercise guidance
- 2 x daily Glucerna® Advance with HMB, Arg and Lys (Abbott)

OUTCOMES MEASURED

Primary - Nutritional status ('MUST');

Secondary - HbA1c, fasting plasma glucose, fat-free mass, phase angle, handgrip strength, chair stand test, sarcopenia diagnosis, quality, quality of life and diabetes distress, GLIM, SGA, SARC-F

Supportive - compliance and acceptability, anthropometrics, demographics, medications, biochemistry parameters, participant and HCP satisfaction surveys

STUDY POPULATION (n=193):

- **MEAN AGE:** 69 years
- 73% male
- 100% high MN risk and T2D
- **MEAN BMI:** 24.2 kg/m²
- **MEAN A1C:** 6.62%
- **MEAN BODY WEIGHT:** 65.6 kg
- **10 YEARS** with T2DM
- 87% oral antidiabetic meds
- 23% insulin use

RESULTS:

56% of patients improved* their nutritional status in just 90 days

*Any improvement from severe to moderate/no malnutrition per GLIM

MALNUTRITION RISK

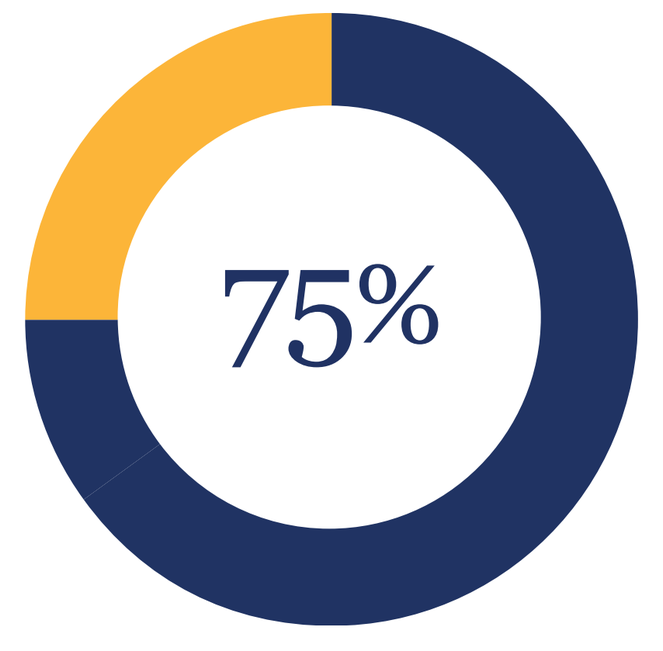
75%

of patients did not experience clinically significant weight loss

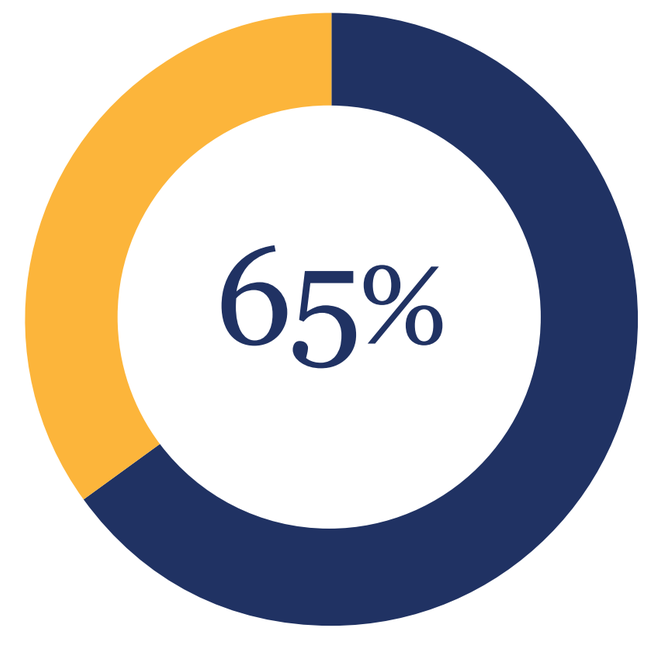
65%

improved their MN risk category (p<0.0001)

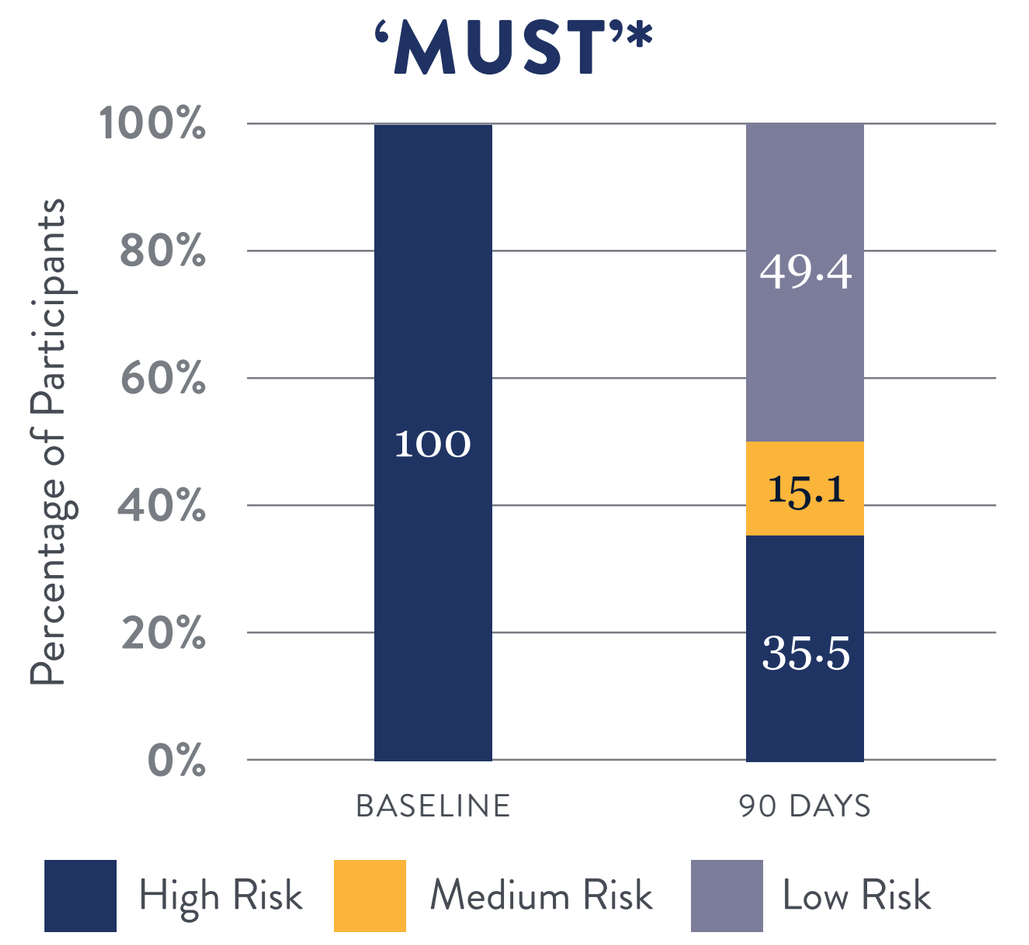
'MUST'*
100%
80%
60%
40%
20%
0%
Percentage of Participants
100
49.4
15.1
35.5
BASELINE
90 DAYS
High Risk
Medium Risk
Low Risk

*p<0.0001 (Bowker's Test)

MALNUTRITION DIAGNOSIS

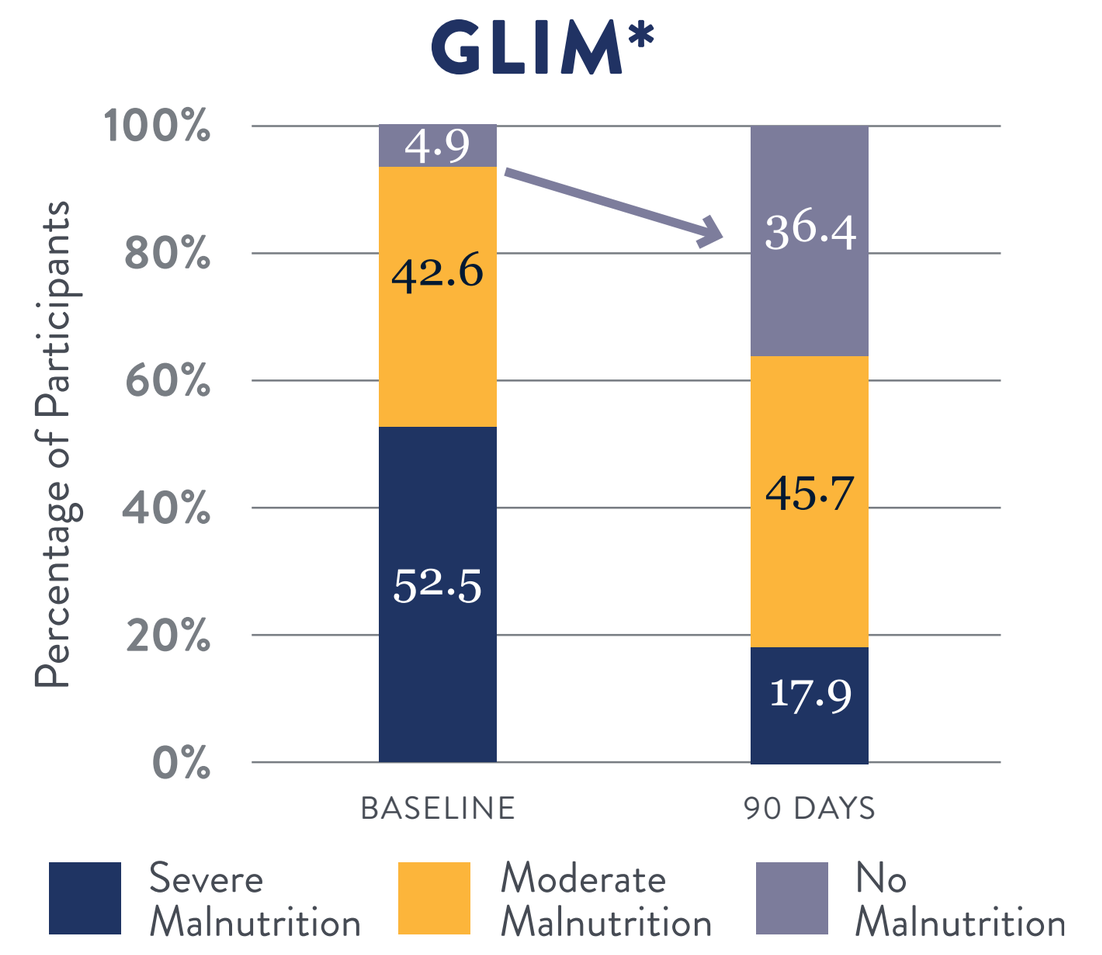
GLIM*
100%
80%
60%
40%
20%
0%
Percentage of Participants
4.9
42.6
52.5
36.4
45.7
17.9
BASELINE
90 DAYS
Severe Malnutrition
Moderate Malnutrition
No Malnutrition

*p<0.0001 (Bowker's Test)

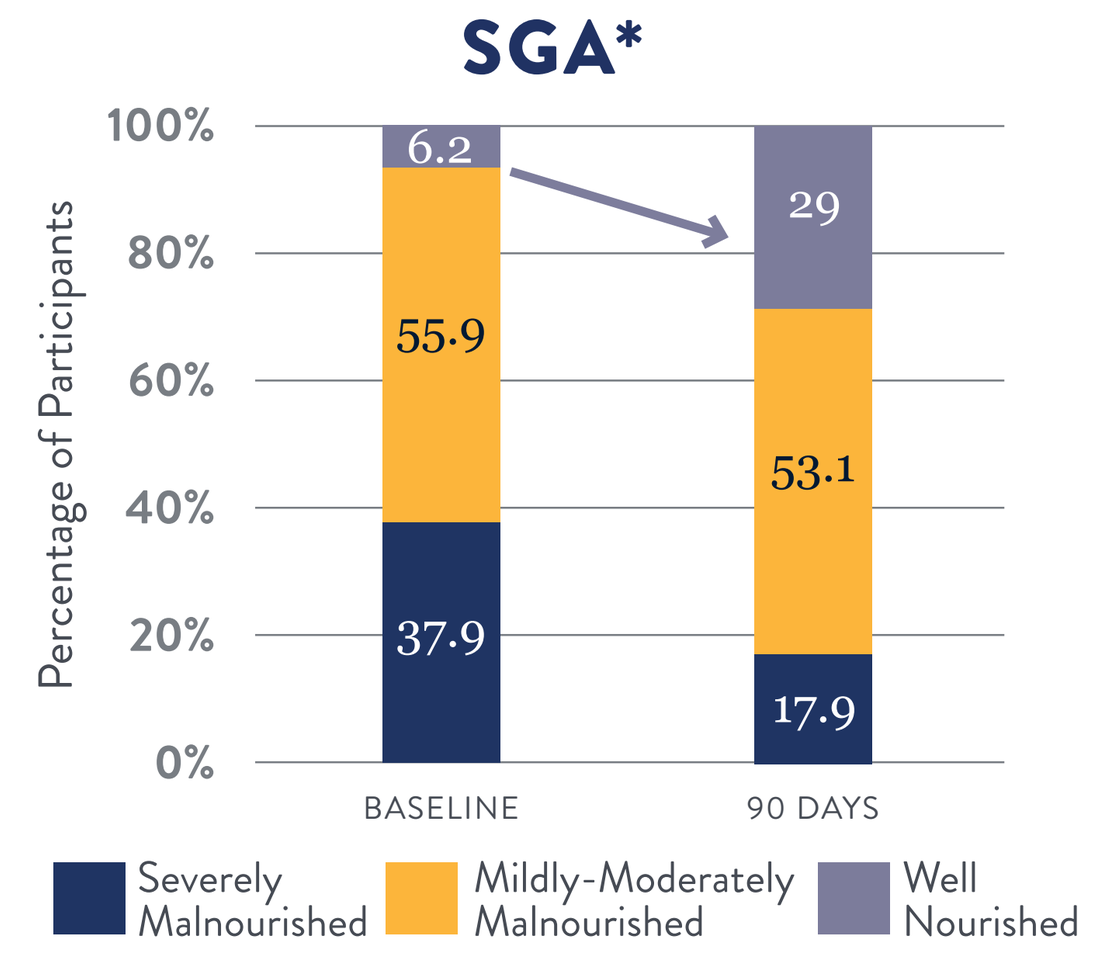
SGA*
100%
80%
60%
40%
20%
0%
Percentage of Participants
6.2
55.9
37.9
29
53.1
17.9
BASELINE
90 DAYS
Severely Malnourished
Mildly-Moderately Malnourished
Well Nourished

*p<0.0001 (Bowker's Test)

Significant improvement in muscle quantity and quality (mean change)

- **+1.3** rises/30s — **IMPROVED CHAIR STAND TEST** (±0.3, p<0.0001)
- **+1.1** kg — **INCREASED FAT-FREE MASS** (±0.4, p=0.0110)
- **+0.5** degrees — **INCREASED PHASE ANGLE** (±0.05, p<0.0001)
- **+2** kg — **INCREASED BODY CELL MASS** (±0.3, p<0.0001)
- **+0.8** kg — **INCREASED** HANDGRIP STRENGTH** SD(±0.4, p=0.049)

**In those patients taking antidiabetic medications (87% of studied population)

Improved Glycemic Control

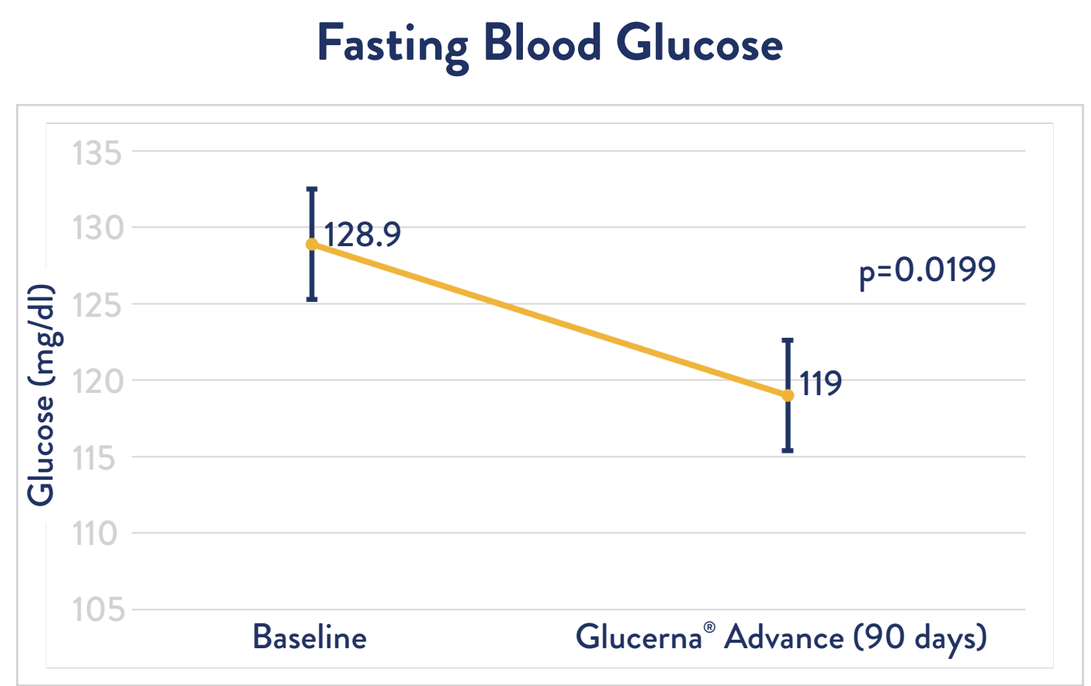
Fasting Blood Glucose
135
130
125
120
115
110
105
Glucose (mg/dl)
128.9
119
p=0.0199
Baseline
Glucerna® Advance (90 days)

IMPROVED FASTING BLOOD GLUCOSE

(change:-9.9±4.2,mg/dL p=0.0199)

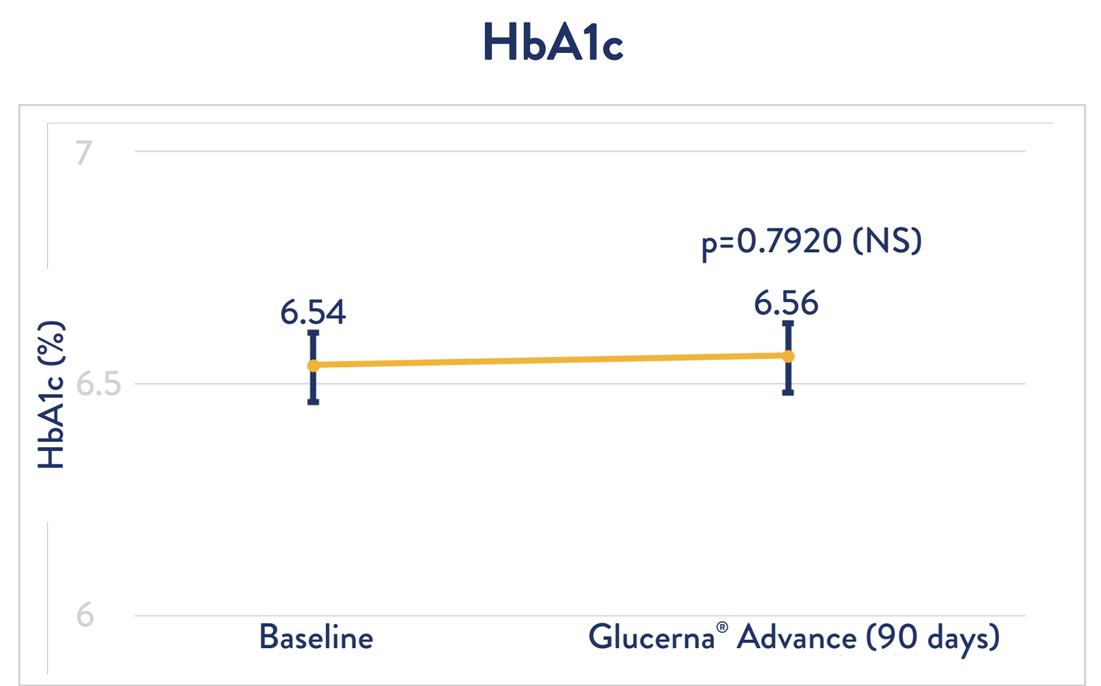
HbA1c
7
6.5
6
HbA1c (%)
6.54
6.56
p=0.7920 (NS)
Baseline
Glucerna® Advance (90 days)

A1C LEVELS REMAINED STABLE

Additional Results

- **32%** increase in Vitamin D levels (From 22.69 to 29.98 ng/mL, p=<0.0001)
- **26%** reduction in proportion of subjects with sarcopenia
- Triglycerydes reduced by **10%** (from 136.5 ± 6.3 to 123.5 ± 4.8 mg/dL, p=<0.001))
- Diabetes distress significantly reduced in just 90 days (by Diabetes distress scale, p=<0.001)
- Quality of life significantly improved (by EQ-ED, p=<0.01)
- Well tolerated in 100% of patients. ~70% of HCPs agreed that the ONS supported their patient's nutritional goals

CONCLUSION:

A 90-day multi-modal nutritional program using Glucerna® Advance with HMB, Lys and Arg:

+ demonstrated meaningful improvements in glycemic control, muscle quantity and quality, cardiometabolic health, quality of life and diabetes distress in people with T2DM and malnutrition.
+ was highly accepted by HCPs and people with T2DM and malnutrition and well suited to routine clinical practice

References

1. Garcia-Almeida JM *et al.*, Effect Of A Nutritional Support Program Including A Diabetes-Specific Nutritional Formula With β-Hydroxy β-Methylbutyrate (HMB) On Nutritional Status And Muscle Health In Malnourished Patients With Diabetes In A Reallife Setting: Preliminary Results Clinical Nutrition Espen 69 (2025) 824–1139. 2. Lopez-Pedrosa JM *et al.*, The Vicious Cycle Of Type 2 Diabetes Mellitus And Skeletal Muscle Atrophy: Clinical, Biochemical, And Nutritional Bases. Nutrients 2024, 16, 172. 3. De Luis Roman D *et al.*, Diabetic Sarcopenia. A Proposed Muscle Screening Protocol In People With Diabetes. Rev In Endo And Metab Dis 2024, Vol 25, 651-661. 4. Manzano M *et al.*, Arginine And Lysine Supplementation Potentiates The Beneficial-Hydroxy ß-Methyl Butyrate (hmb) Effects On Skeletal Muscle In A Rat Model Of Diabetes. Nutrients 2024, 15, 4796.

Disclosures

M Camprubi Robles, G Guzman Rolo, G. E. Baggs are employees of Abbott. P. Suryawanshi is an employee of Statistical Services, Cognizant Technologies Solution Pvt. Ltd. Financial support for this study was provided by Abbott.

Funding: this study was funded by Abbott laboratories.

ABBOTT